Rapid Onset of Efficacy of XEN1101, a Novel Potassium Channel Opener, in Adults with Focal Epilepsy: Results from a Phase 2b Study (X-TOLE)

Christopher Kenney,[1] Jacqueline French,[2] Roger Porter,[3] Emilio Perucca,[4] Martin Brodie,[5] Michael A. Rogawski,[6] Cynthia Harden,[1] Constanza Luzon Rosenblut,[1] Jenny Qian,[1] Jennifer Leung,[1] Gregory N. Beatch[1]

[1]Xenon Pharmaceuticals Inc., Burnaby, BC, Canada; [2]New York University Comprehensive Epilepsy Center, New York, NY; [3]University of Pennsylvania, Philadelphia, PA; [4]University of Melbourne and Monash University, Melbourne, Australia; [5]University of Glasgow Department of Medicine and Therapeutics, Western Infirmary, Glasgow, Scotland; [6]School of Medicine, University of California, Davis, Sacramento, CA

INTRODUCTION

- XEN1101 is a novel, potent, selective KCNQ2/3 (Kv7.2/7.3) potassium channel opener being developed for the treatment of focal onset seizures (FOS). Evaluation of XEN1101 in Phase 1 clinical studies determined that XEN1101 had a long half-life, which supported a once-daily dosing schedule and suggested that a titration/tapering regimen was not needed.
- Despite the availability of several new antiseizure medications (ASMs), approximately 30% of patients still experience uncontrolled seizures. With each ASM failure, the likelihood of achieving seizure control with each subsequent ASM regimen decreases.[1] Changes to an ASM regimen are often frequent due to lack of efficacy or intolerability.
- Very few ASMs can be initiated at both a therapeutic and well-tolerated dose, due to side effects or pharmacokinetic properties. As a result of the diverse interindividual responses to ASMs, titration and tapering for several weeks may be required when initiating or ending a therapy. A "start low, go slow" titration approach is used to avoid severe adverse effects.[2] However, a titration period can result in suboptimal ASM dosing which may lead to breakthrough seizures.
- In the recently completed X-TOLE Phase 2b clinical study, the clinical efficacy, safety, and tolerability of XEN1101 administered as adjunctive treatment was evaluated in adults with FOS. In this analysis we further investigated the time to onset of action by evaluating the change in weekly FOS frequency and the number of responders achieving ≥50% reduction (RR50) at Week 1.

METHODS

Key Inclusion Criteria

- Patients aged 18-75 years (inclusive) with an International League Against Epilepsy [ILAE][3] diagnosis of focal epilepsy (≥2 years).
- ≥4 countable focal seizures per month, recorded on an eDiary during a planned 8-week baseline period, while receiving stable treatment with 1-3 ASMs.

Study Design

- Subjects were randomized, for an 8-week, double-blind phase to one of three active treatment groups or placebo (**Figure 1**) in a 2:1:1:2 ratio (XEN1101 25 mg: 20 mg: 10 mg: placebo).
- Monthly (28 days) FOS frequency was calculated as the total number of FOS reported × 28 / the total number of days seizure information was available.
- Weekly (7 days) FOS frequency was defined as the total number of seizures every 7 days. For the double-blind period (DBP) the weekly period started on the randomization date and assumed 7-day weeks until treatment end date.
- A prespecified weekly assessment of seizure frequency was conducted followed by a post hoc statistical pair-wise comparison between placebo and each treatment group.
- Monthly and weekly response (RR50) was computed as those subjects having achieved ≥50% reduction in focal seizure frequency from baseline, based on percent change from baseline in focal seizure frequency (based on countable seizure types 1-4).

Table 1. Baseline Demographic and Clinical Characteristics (Safety Population)

	Placebo (N=114)	XEN1101 10 mg (N=46)	XEN1101 20 mg (N=51)	XEN1101 25 mg (N=114)	TOTAL (N=325)
Age at study entry, years					
Mean (SD)	42.9 (13.7)	40.0 (12.1)	41.7 (13.6)	38.7 (13.1)	40.8 (13.3)
Gender					
Female, n (%)	61 (53.5)	27 (58.7)	26 (51.0)	54 (47.4)	168 (51.7)
Male, n (%)	53 (46.5)	19 (41.3)	25 (49.0)	60 (52.6)	157 (48.3)
Baseline monthly focal onset seizure frequency					
Mean (SD)	27.3 (38.5)	35.5 (40.9)	29.0 (42.0)	23.5 (30.4)	27.4 (36.9)
Median	13.4	17.4	14.5	12.8	13.5
Background antiseizure medication use					
1, n (%)	12 (10.5)	4 (8.7)	2 (3.9)	11 (9.6)	29 (8.9)
2, n (%)	46 (40.4)	18 (39.1)	20 (39.2)	47 (41.2)	131 (40.3)
3, n (%)	56 (49.1)	24 (52.2)	29 (56.9)	56 (49.1)	165 (50.8)

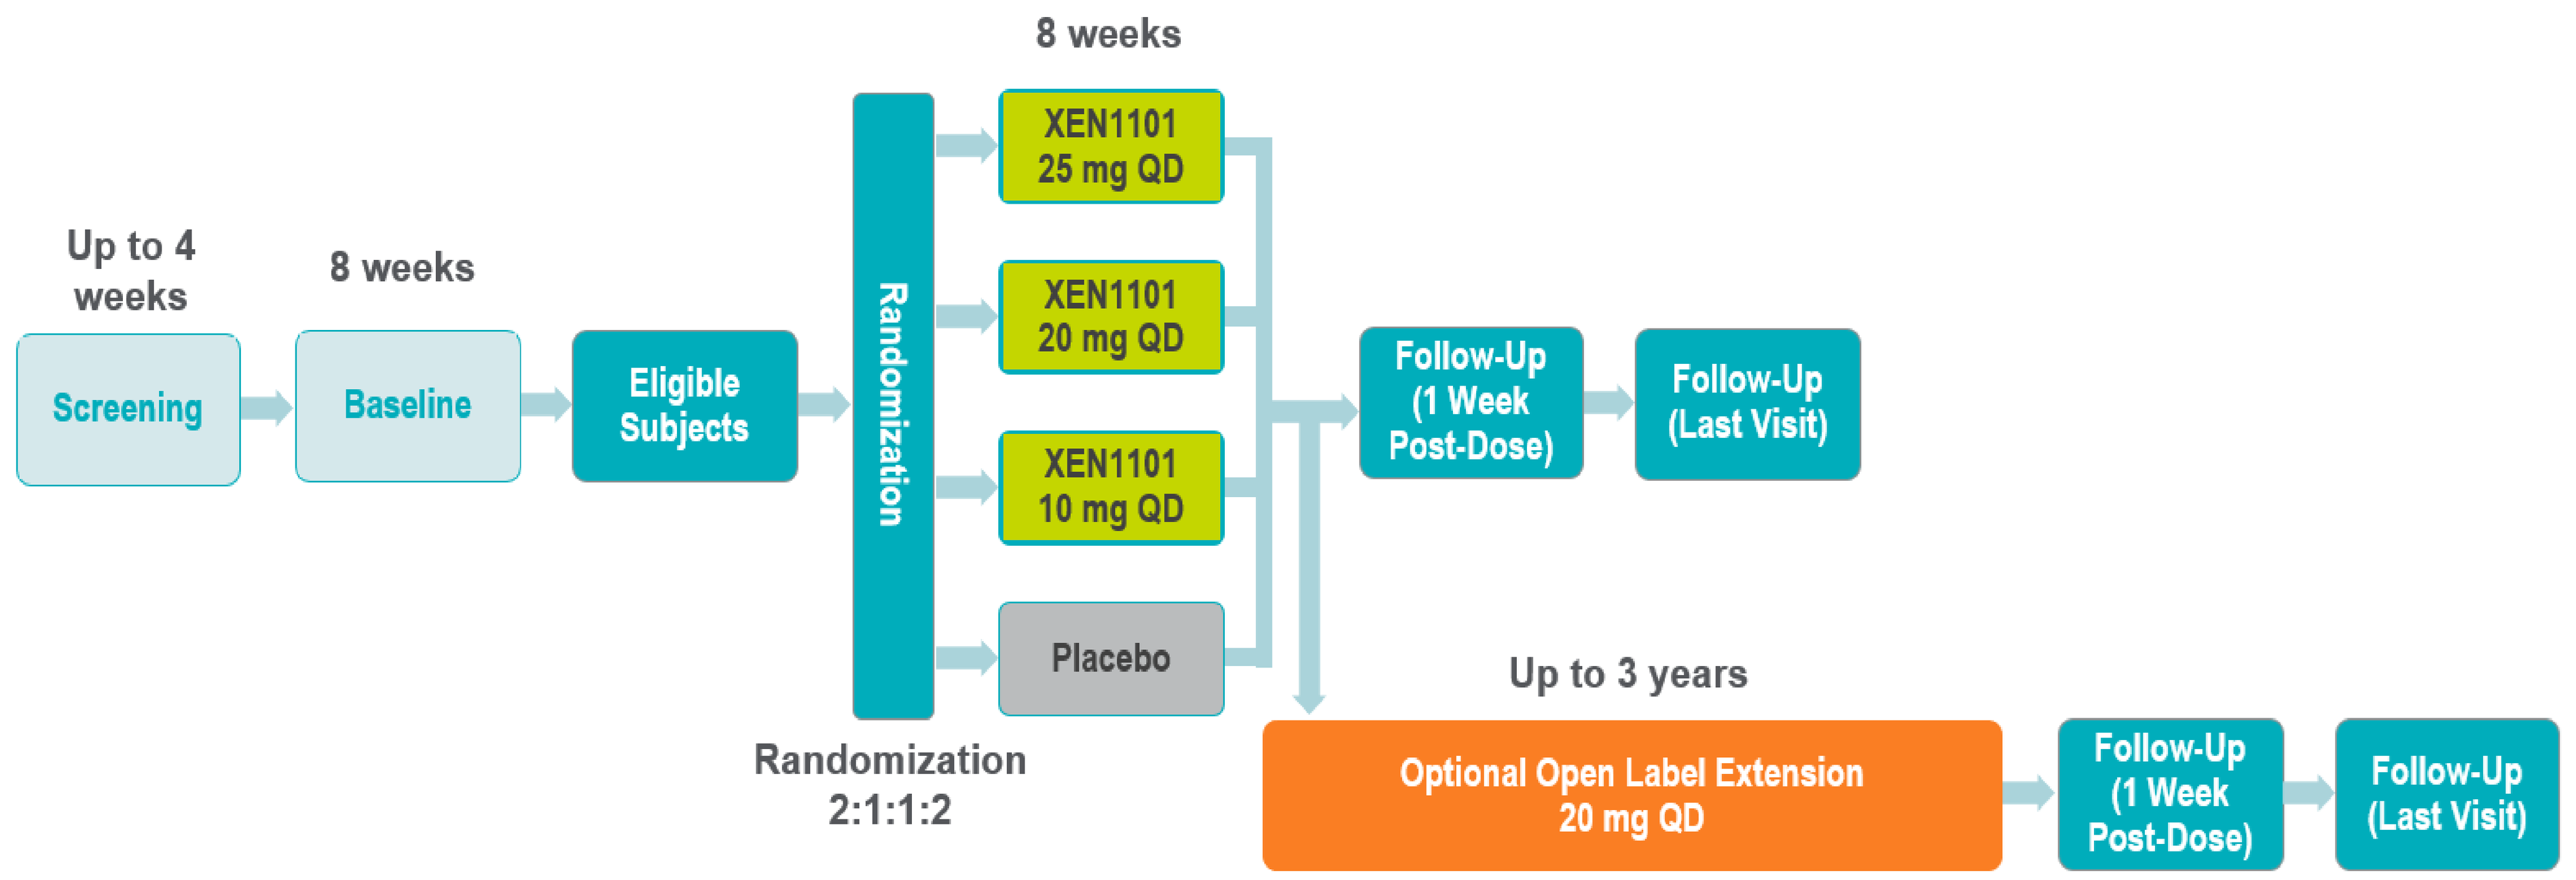
Up to 4 weeks
Screening
8 weeks
Baseline
Eligible Subjects
Randomization
Randomization 2:1:1:2
8 weeks
XEN1101 25 mg QD
XEN1101 20 mg QD
XEN1101 10 mg QD
Placebo
Follow-Up (1 Week Post-Dose)
Follow-Up (Last Visit)
Up to 3 years
Optional Open Label Extension 20 mg QD
Follow-Up (1 Week Post-Dose)
Follow-Up (Last Visit)

Figure 1 X-TOLE Study Design

RESULTS

- Baseline demographic and clinical characteristics are shown in **Table 1**.
- XEN1101 demonstrated a dose-dependent reduction from baseline in median monthly FOS frequency (**Figure 2A**) of -33.2% (p=0.035, n=46), -46.4% (p<0.001, n=51), and -52.8% (p<0.001, n=112) in the 10 mg, 20 mg, and 25 mg groups, respectively, compared to placebo (-18.2%, n=114).
- Rapid onset of efficacy of XEN1101 was seen at Week 1 (**Figure 2B**), with a dose-dependent reduction from baseline in median weekly FOS frequency of -39.1% (p<0.01, n=46), -41.5% (p=0.04, n=50) and -55.4% (p<0.001, n=110) in the 10 mg, 20 mg, and 25 mg groups, respectively, compared to placebo (-20.2%, n=114).

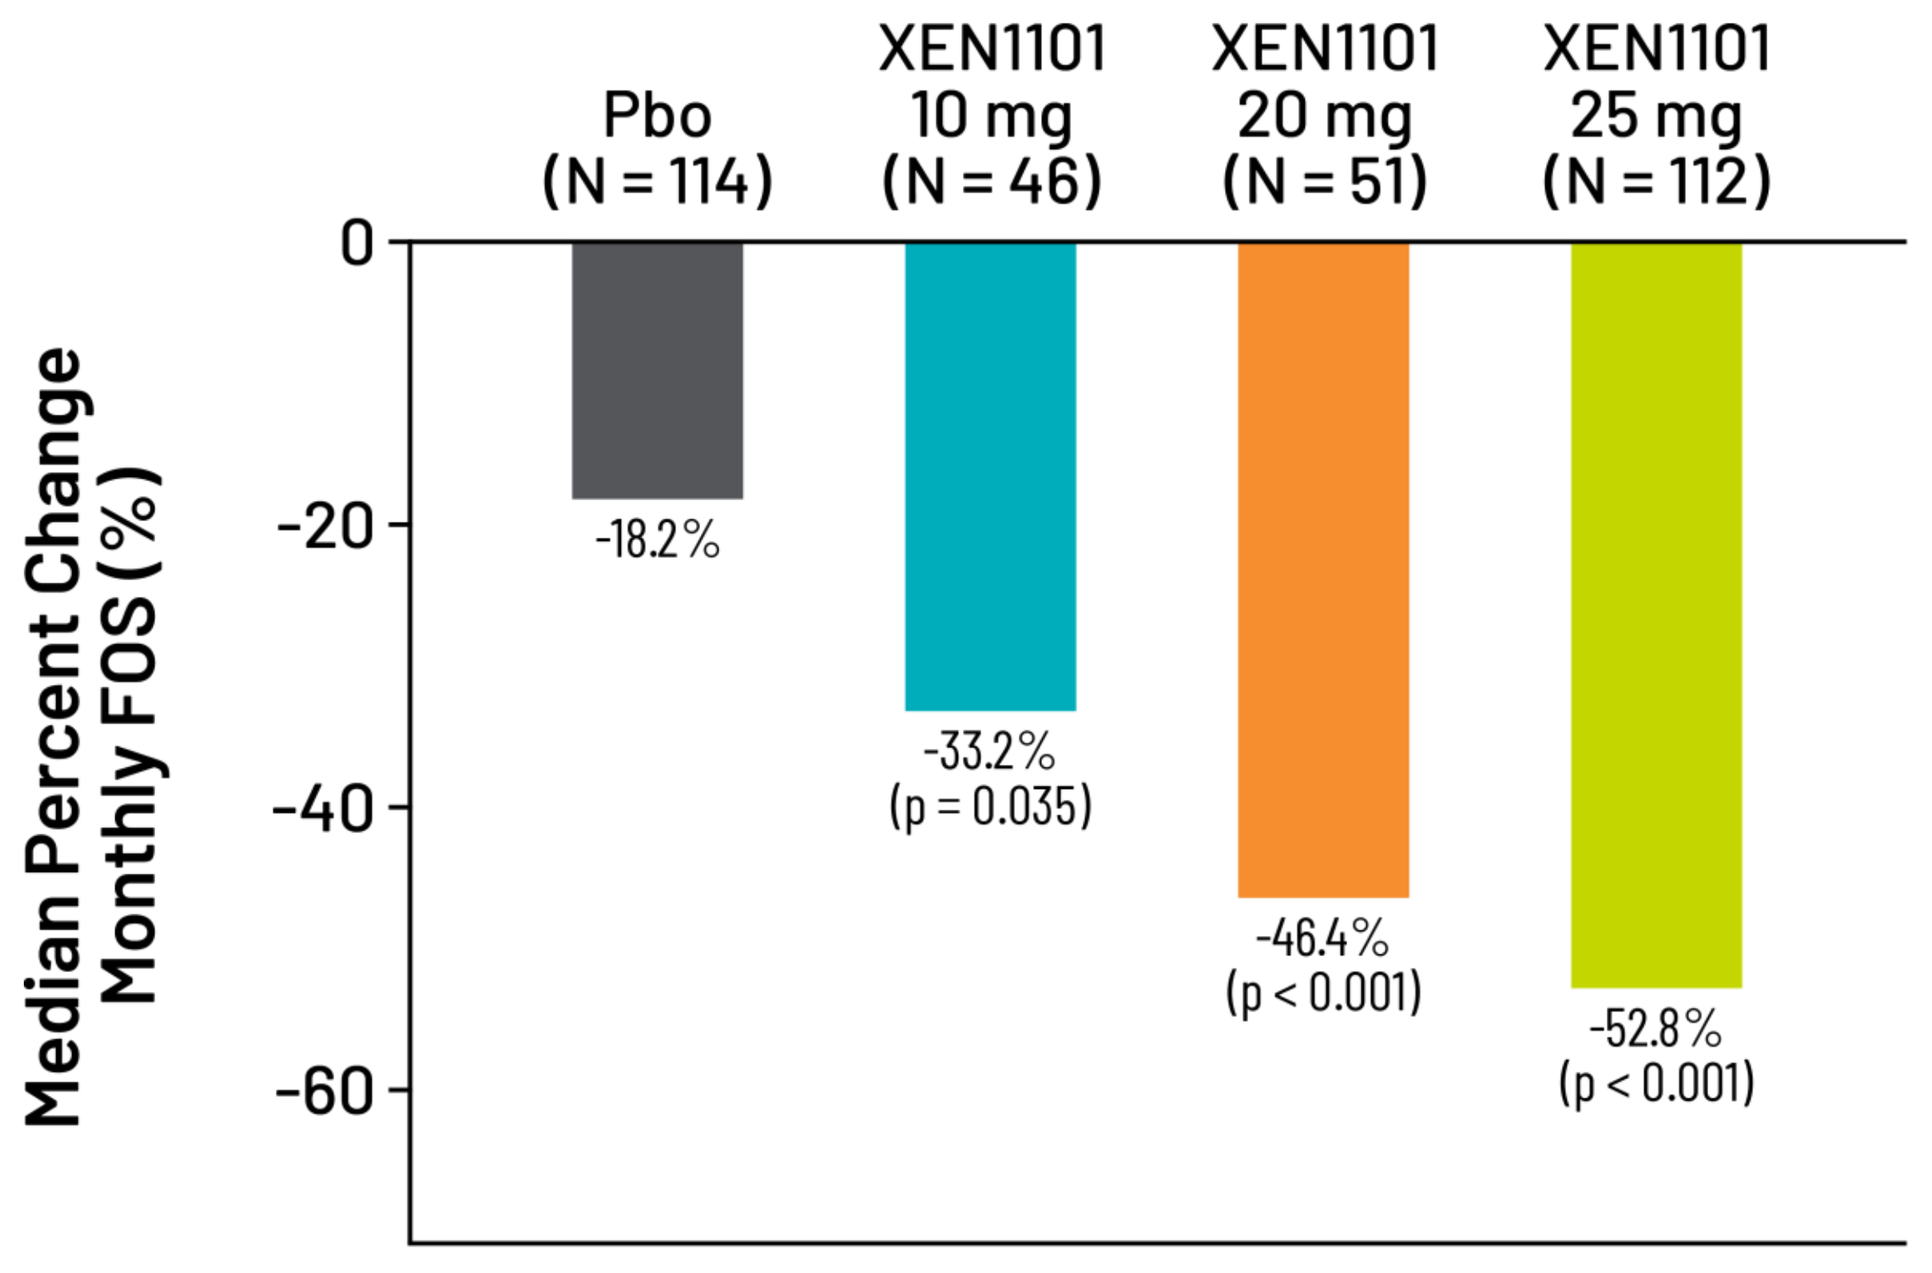
Pbo (N = 114)
XEN1101 10 mg (N = 46)
XEN1101 20 mg (N = 51)
XEN1101 25 mg (N = 112)
Median Percent Change Monthly FOS (%)
0
-20
-40
-60
-18.2%
-33.2% (p = 0.035)
-46.4% (p < 0.001)
-52.8% (p < 0.001)

Figure 2A Overall Median Percent Change from Baseline in Monthly Focal Onset Seizure (FOS) Frequency in the Double-Blind Period

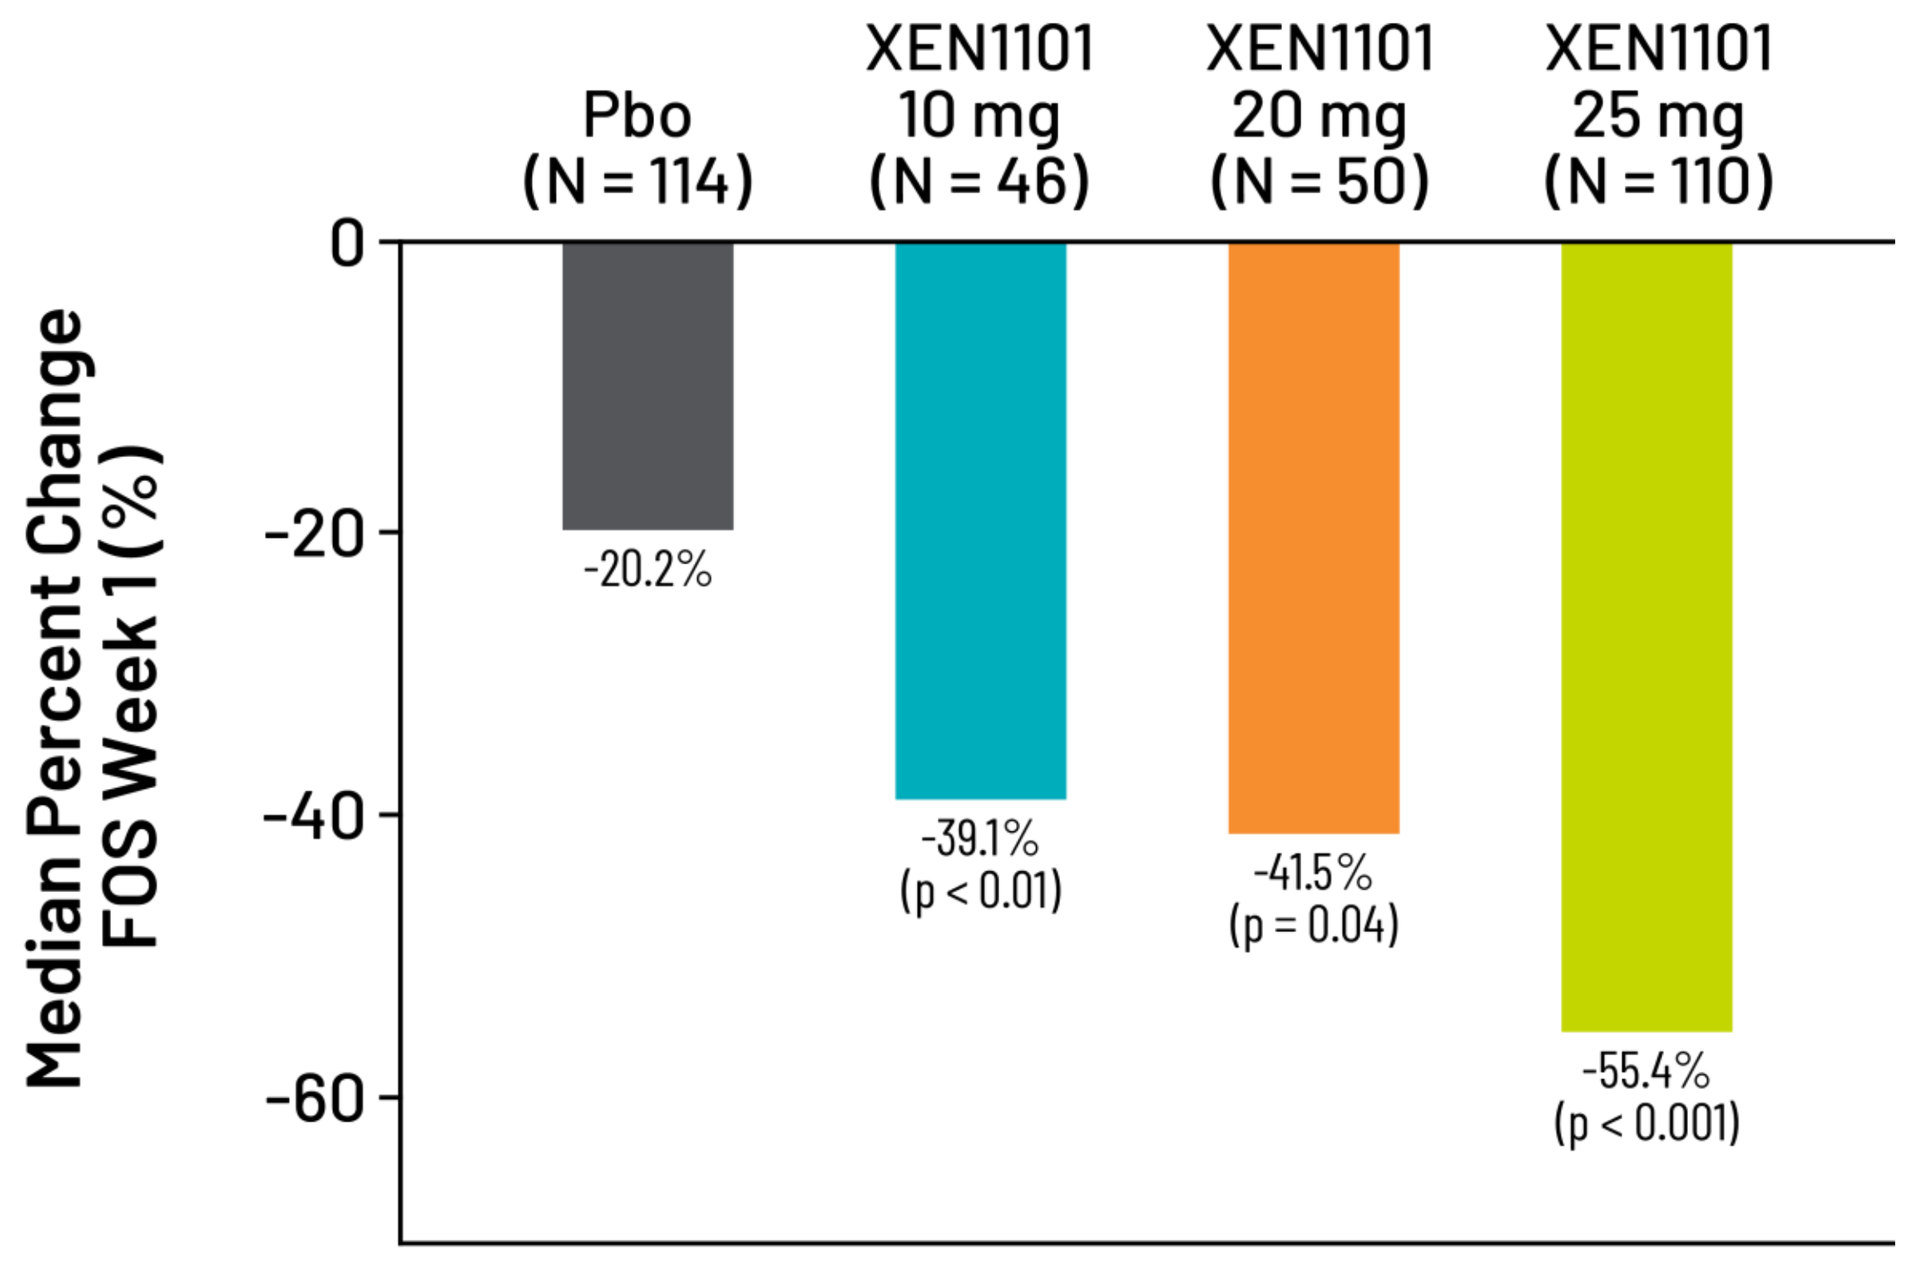
Pbo (N = 114)
XEN1101 10 mg (N = 46)
XEN1101 20 mg (N = 50)
XEN1101 25 mg (N = 110)
Median Percent Change FOS Week 1 (%)
0
-20
-40
-60
-20.2%
-39.1% (p < 0.01)
-41.5% (p = 0.04)
-55.4% (p < 0.001)

Figure 2B Median Percent Change from Baseline in Weekly Focal Onset Seizure (FOS) Frequency During Week 1 of the Double-Blind Period

- XEN1101 demonstrated dose-dependent increases in the number of responders with ≥50% reduction in monthly (**Figure 3A**) and weekly (**Figure 3B**) FOS frequency.
- Median percent change in FOS frequency at Week 1 shows rapid and dose-dependent seizure suppression.
- Efficacy of XEN1101 was sustained throughout the 8-week DBP for the 20 mg and 25 mg groups (**Figure 4**).

Safety

- The most common treatment emergent adverse events (TEAEs) leading to discontinuation across XEN1101 groups were dizziness (4.7%), balance disorder (2.4%), dysarthria (1.9%), and gait disturbance (1.9%). There were no idiosyncratic immunologic adverse reactions when XEN1101 was initiated at a therapeutic dose.
- For the 25 mg treatment group, 21 subjects (18.4%) dose-reduced due to a TEAE. Of these, only 3 subsequently discontinued treatment permanently. This indicates that dose reduction prevented early termination when it was applied.

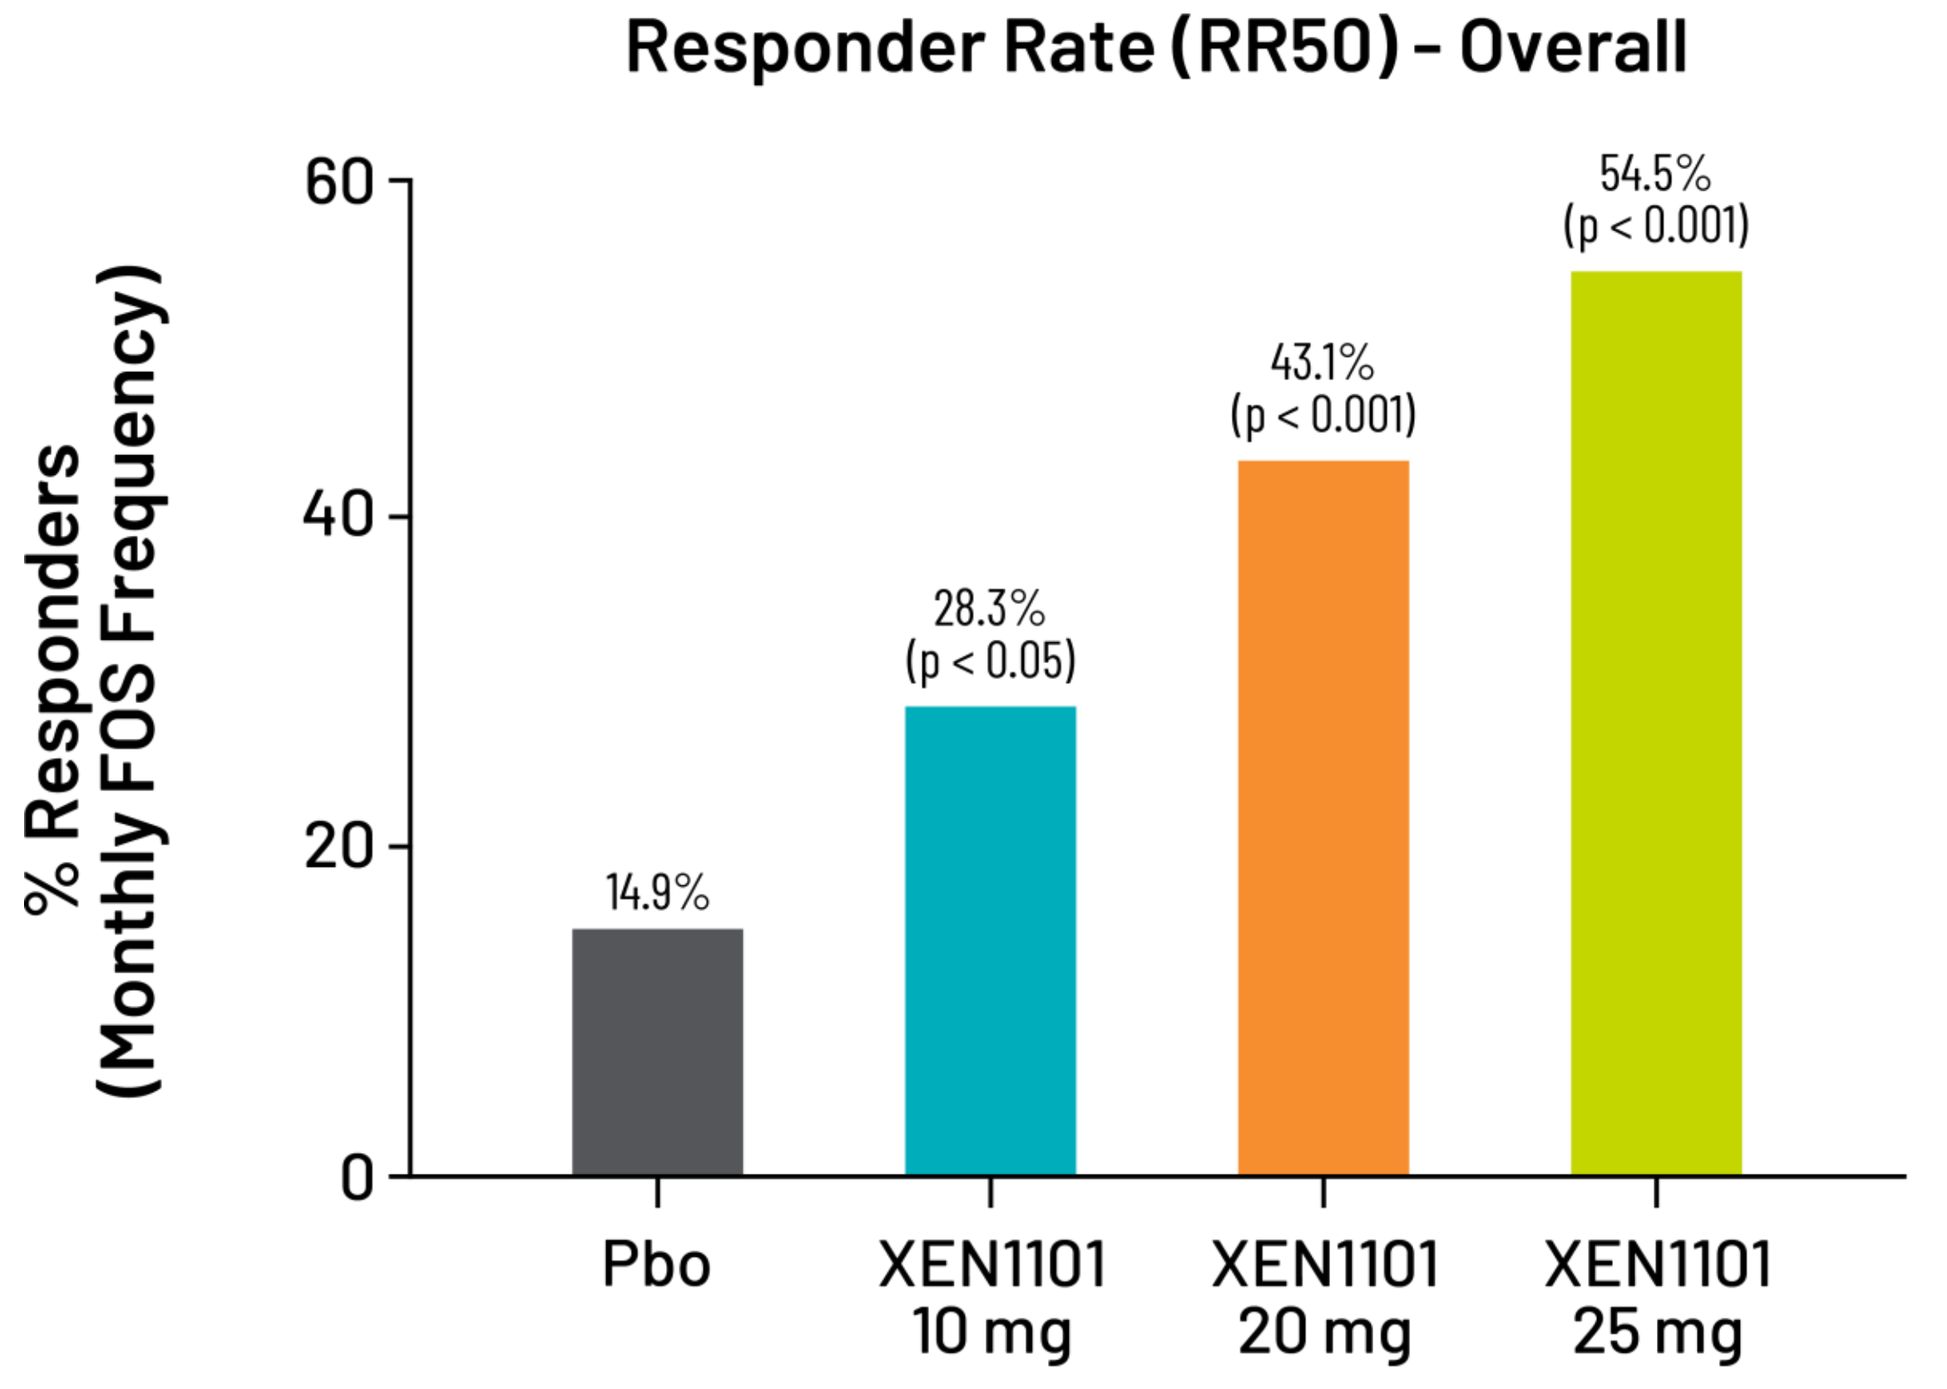
Responder Rate (RR50) - Overall
% Responders (Monthly FOS Frequency)
60
40
20
0
14.9%
28.3% (p < 0.05)
43.1% (p < 0.001)
54.5% (p < 0.001)
Pbo
XEN1101 10 mg
XEN1101 20 mg
XEN1101 25 mg

Figure 3A Overall Responders (RR50) Based on Percent Change from Baseline in Monthly Focal Onset Seizure (FOS) Frequency in the Double-Blind Period

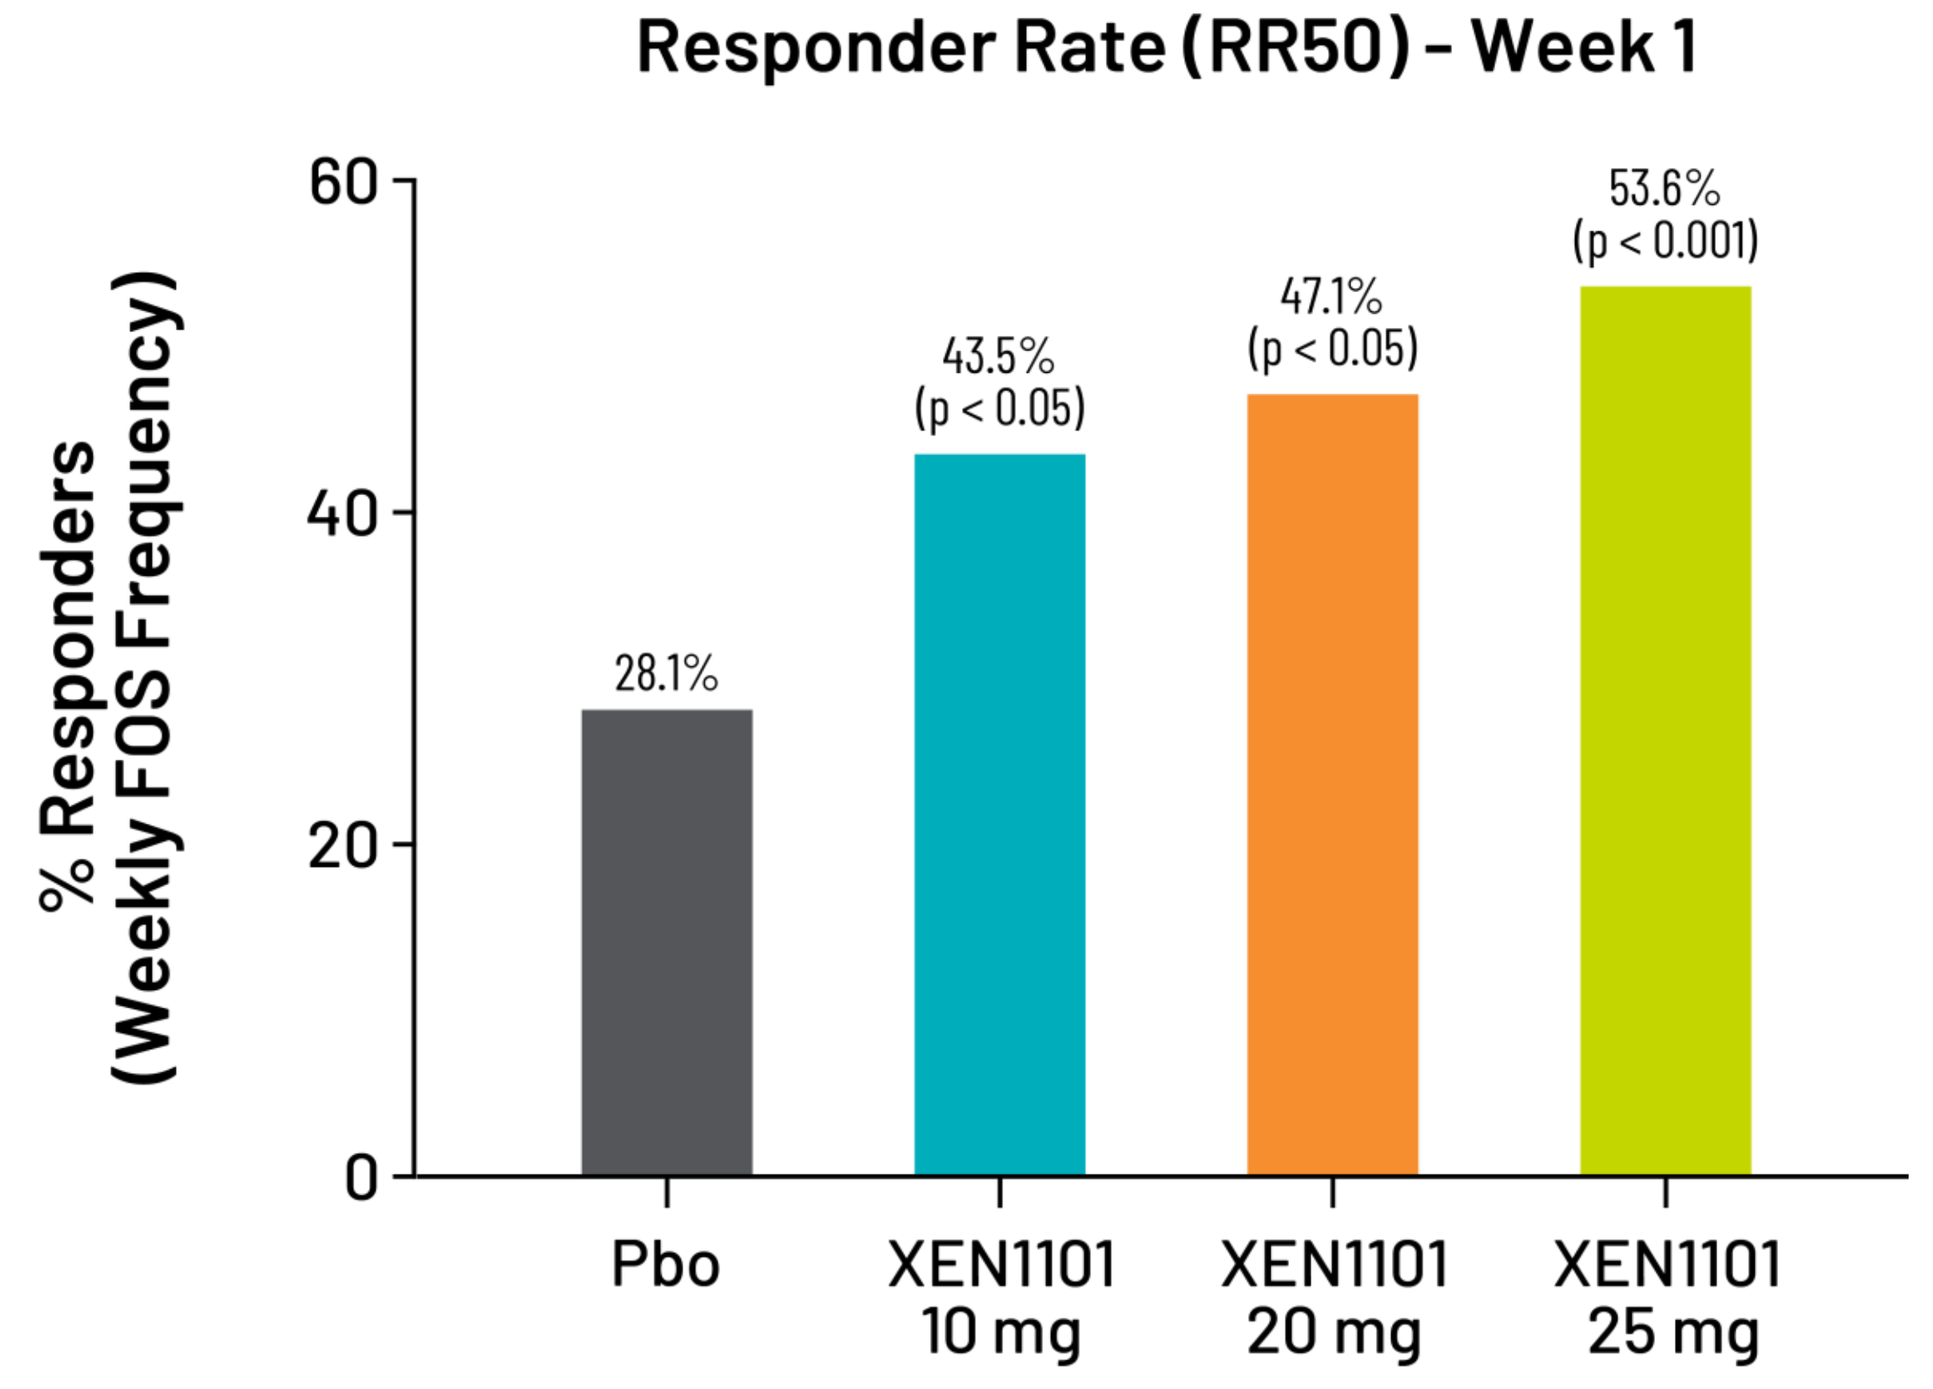
Responder Rate (RR50) - Week 1
% Responders (Weekly FOS Frequency)
60
40
20
0
28.1%
43.5% (p < 0.05)
47.1% (p < 0.05)
53.6% (p < 0.001)
Pbo
XEN1101 10 mg
XEN1101 20 mg
XEN1101 25 mg

Figure 3B Responders (RR50) Based on Percent Change from Baseline for Week 1 in Weekly Focal Onset Seizure (FOS) Frequency in the Double-Blind Period

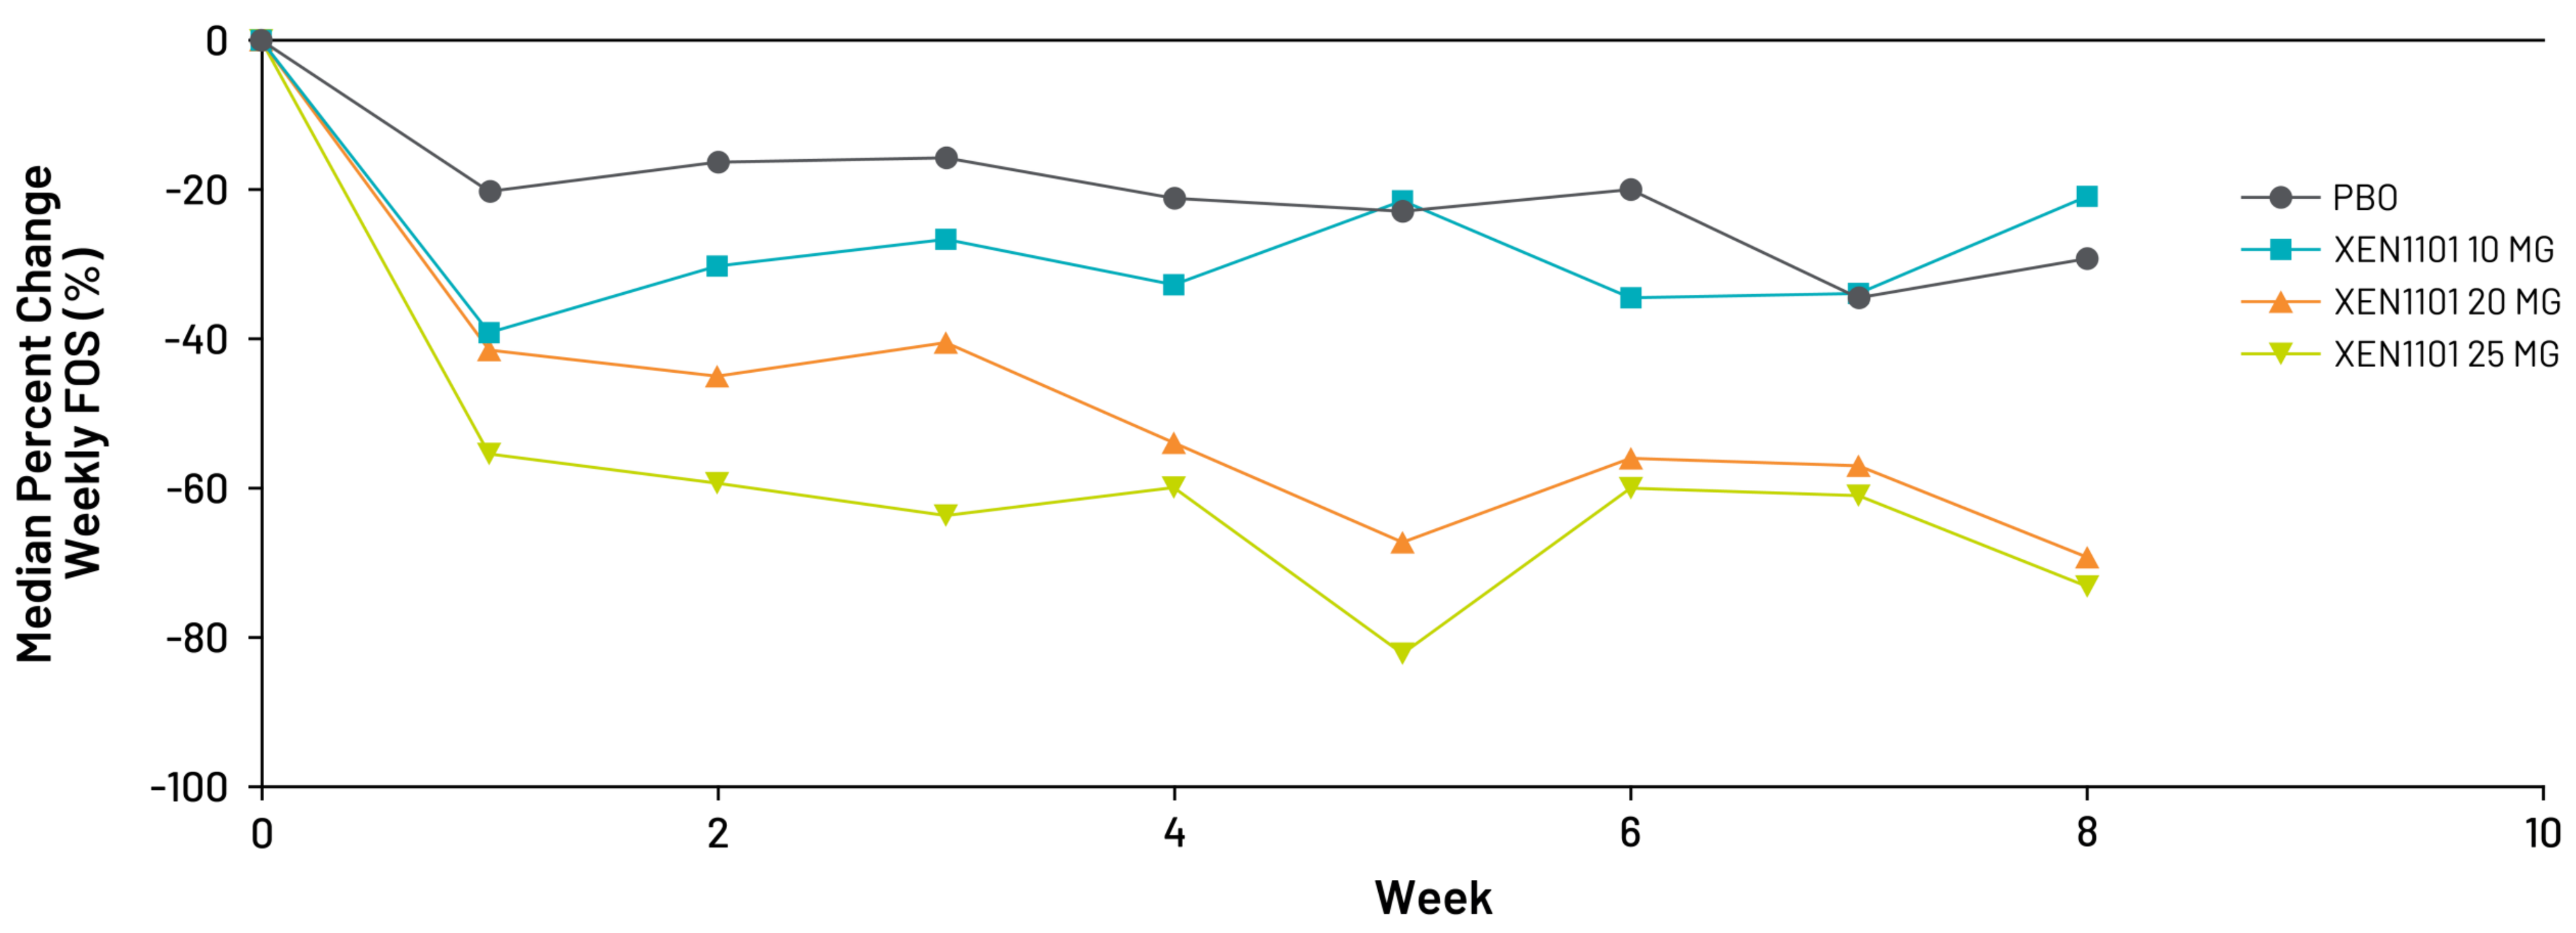
Median Percent Change Weekly FOS (%)
0
-20
-40
-60
-80
-100
0
2
4
6
8
10
Week
PBO
XEN1101 10 MG
XEN1101 20 MG
XEN1101 25 MG

Figure 4 Median Percent Change from Baseline in Weekly Focal Onset Seizure (FOS) Frequency for the Double-Blind Period (Modified Intent-To-Treat Population)

CONCLUSIONS

- X-TOLE met the primary and key secondary efficacy endpoints with XEN1101 demonstrating a statistically significant, dose-dependent reduction from baseline in monthly FOS frequency compared to placebo.
- The rapid onset of efficacy for XEN1101 was associated with starting at an effective, therapeutic and well-tolerated dose. There was a marked reduction in median FOS frequency within 1 week for all doses compared with placebo.
- The rapid onset of efficacy after 1 week and sustained efficacy of XEN1101 remain to be confirmed in Phase 3 clinical trials. XEN1101 may offer a compelling option for patients seeking an adjunctive therapy that quickly provides seizure reduction.

References

1. Chen et al. *JAMA Neurology*. 2018;75(3):279-286.
2. Seiden et al. *Epilepsy & Behavior*. 2022;128:108517.
3. Fisher et al. *Epilepsia*. 2017;58(4):522-530.

Poster #451 | European Epilepsy Congress | July 9-13, 2022 | Geneva, Switzerland

XENON